Abstract #2191

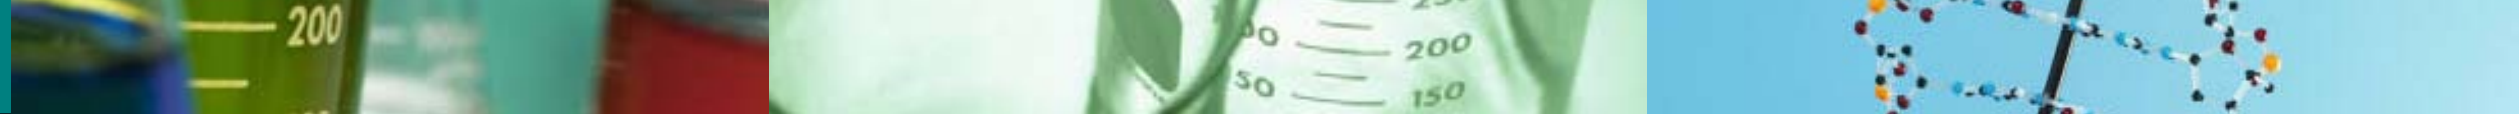

Anti-metastatic activity of ARQ 197, a highly selective oral small molecule inhibitor of c-Met, in experimental metastatic models of colon cancer

ARQULE

BOSTON BIOMEDICAL

Youzhi Li[2], Wei Zhou[2], Dongshu Chen[2], Wei Li[2], Zhiwei Jiang[2], Taiping Chen[1], David S. Leggett[2], Chiang J. Li[1,2]

[1]ArQule Inc., 19 Presidential Way, Woburn, MA 01801, [2]Boston Biomedical, Inc., 333 Providence Highway, Norwood, MA 02062

ABSTRACT

The oncogene c-Met is a key molecular regulator of the invasive and metastatic growth of cancer cells, the ultimate cause of death from malignancy. Most colorectal cancer patients develop metastases to the liver or lung during the course of the disease. They are associated with poor prognosis, resulting in the death of more than 80% of patients. Recently, we have discovered ARQ 197, a highly selective small molecule inhibitor of c-Met. ARQ 197 kills a wide range of cancer cells by selectively targeting c-Met *in vitro* and has shown potent anti-tumor activity against several types of xenografted human tumors in mice. In the present study, we investigated anti-metastatic effects of ARQ 197 using our liver metastatic model of colon carcinoma in mice. In this model, colon cancer cells were implanted orthotopically and liver metastasis can be formed spontaneously. Our data show that ARQ 197 was well tolerated and displayed a significant inhibition of liver metastasis. In untreated control group, spontaneous liver metastasis were formed in nearly 80% of mice. The metastatic lesions were confirmed histologically. Treatment with ARQ 197 significantly reduced or prevented liver metastases with less than 10% of mice exhibiting liver metastases. ARQ 197 treatment also resulted in a significant reduction of visually detectable pulmonary metastases in SCID mice induced by i.v. injection of HT29 human colon carcinoma cells. No significant adverse effects were observed based on body weight and the general appearance. In summary, ARQ 197, a highly selective oral small molecule c-Met inhibitor, potently blocked metastasis of human colon carcinoma in an animal model.

Introduction

- Colorectal carcinoma (CRC) is the third leading cause of cancer-related deaths worldwide. Most colorectal cancer patients develop metastases to the liver or lung during the course of the disease. They are associated with poor prognosis, resulting in the death of more than 80% of patients. Therefore, two metastatic colorectal tumor models were used in the current study. Intrasplenic-nude mouse model system (ISMS model) is especially appropriate for studies of the malignant behavior of colorectal carcinomas, as this technique commonly produces experimental metastases in the liver, and the pulmonary metastases in SCID mice systemically disseminated by intravenous injection of tumor cells, the most common site of metastases of this carcinoma.
- c-Met is a transmembrane tyrosine kinase receptor for Hepatocyte growth factor/scatter factor (HGF/SF) encoded by the *c-met* proto-oncogene. Its role in cancer development is definitively established as it is inappropriately expressed in many types of human cancer. It plays key roles in controlling multiple signal transduction pathways involved in cell growth, cell survival, angiogenesis, invasion and metastasis. Therefore, c-met is a highly compelling therapeutic target for human cancer.
- ARQ 197 has a broad-spectrum activity against cancer cells and selectively and potently inhibits c-Met. In this study, we established two experimental metastasis models and investigated the anti-metastasis role of ARQ 197 in these models. We report here that ARQ 197 was well tolerated and effectively inhibited the colon cancer metastases in both experimental and spontaneous metastatic animal models.

Materials & Methods

➤ **Animals**.

All experiments were carried out using female 5- 6week old Athymic NCr Nu/Nu mice or SCID mice (Charles River Laboratory, Wilmington, MA). Animals were maintained in microisolation cages and maintained in accordance with the guiding principles of ArQule's Institutional Animal Care and Use Committee throughout the studies

➤ **Intrasplenic-nude mouse model system (ISMS model)**

In brief, the mice were anesthetized and under aseptic conditions an incision was made in the left flank to expose the spleen. One million tumor cells in 0.1 ml PBS were injected under the spleen capsule using a 27-gauge needle. The spleen was replaced in the peritoneal cavity and the incision was closed. The mice were sacrificed when moribund or 5 weeks after the injection. The spleen, liver, and lungs were removed and examined, and the number of tumor lesions was recorded.

➤ **Drug Administration.**

The prepared test and control substances were administered via oral administration with either vehicle control or ARQ 197 formulated in PEG400:20% Vitamin E TPGS (60:40) at 30 mg/ml. treated daily (5 consecutive days, followed by a 2-day dosing holiday) for four week period.

➤ **The *intravenous injections* Model**.

The metastatic colonization potential of the HT-29 tumor cells was studied in SCID mice injected intravenously (i.v.) with 2 x 10^6 viable cells in 0.2 ml PBS into the lateral tail vein. Mice were randomized into two groups and treated from day 4 to day 32. The mice were sacrificed on day 38 for autopsy and lung staining. After ink staining, the tumor colonies on the lung surface appeared white, whereas the normal lung tissue appeared black.

➤ **Histology**

Xenografted tumor tissue and organs with suspected occult metastases were fixed in 10 percent buffered formalin, embedded in paraffin, sectioned and stained with hematoxylin and eosin.

➤ **Statistical analyses**.

Statistical analyses in xenograft studies were performed comparing test agent to control groups using a paired Student t-test.

RESULTS

I. Anti-metastatic effect of ARQ 197 on spontaneous liver metastasis ISMS model.

Briefly, Mice were divided into 2 groups; a control group given vehicle (n=9), a group receiving 300 mg/kg ARQ 197 (n=10), were administered orally by gavages for 5 days/week and for 4 weeks from day 4th after *i.s.* injection. Mice were sacrificed on day 34 after *i.s.* injection. The numbers of liver metastases and metastatic liver tumors were estimated histologically and macroscopically. In the vehicle group, spontaneous liver metastasis were formed in nearly 80% of mice. The metastatic lesions were confirmed histologically. Treatment with ARQ 197 significantly reduced or prevented liver metastasis with less than 10% of mice with liver metastasis. These data are illustrated in Figs. A, B & Table C.

A. Representative of *in situ* photographs:

Control @ Day 34

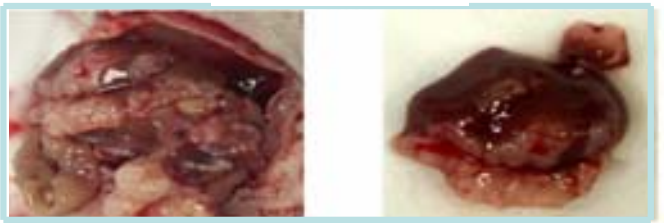

ARQ 197 @ Day 34

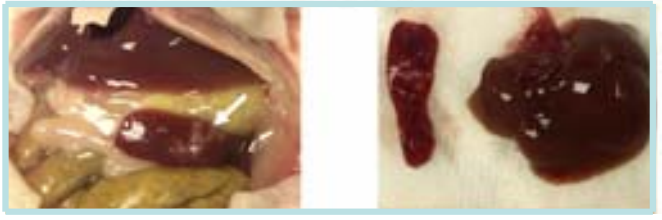

B. Representative of hispathological slides (H&E staining)

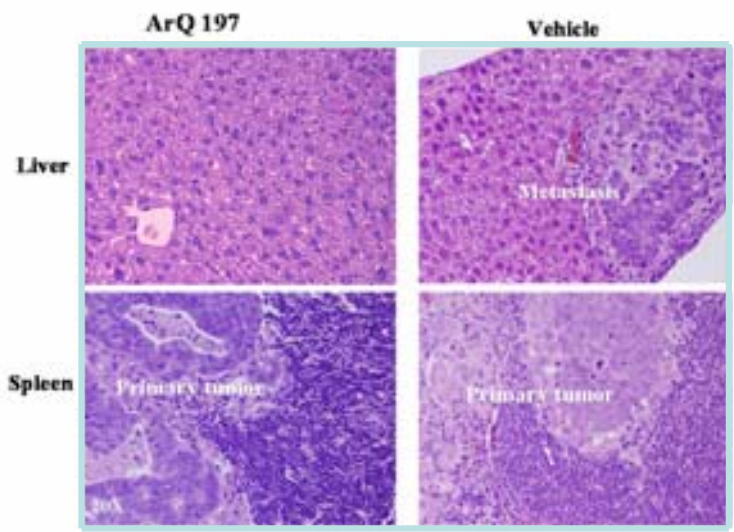

ArQ 197
Vehicle
Liver
Spleen
Metastasis
Primary tumor
Primary tumor

Fig.B. The metastatic lesions were confirmed histologically. Massive metastasis was observed in most mice of the vehicle group, but not in mice of the ARQ 197 treated group.

C. Tumorigenicity and production of metastases by HT-29 cells injected in the spleens of nude mice.

	Control	Incidence	ArQ 197 Treated	Incidence
Spleen	9 Primary Cancer / 9 cases	100%	5 Primary Cancer/ 10 cases	50.00%
			4 Tumor Cell, 1 small foci T/10	50%
Liver	7 Metastasis / 9 cases	77.80%	1 Metastasis /10 cases	10%
			1 Small foci Tumor/10	10%

Table C. demonstrates the effects of vehicle and ARQ 197on the number of liver metastatic foci. In the vehicle group 7/9 (77.8%) had metastatic foci, 300 mg/kg ARQ 197 significantly inhibited the number of metastatic foci 1/10 (10%) compared to the control

II. Anti-metastatic effect of ARQ 197 on experimental metastasis following *intravenous* injection

Briefly, Mice were divided into 2 groups; a control group given vehicle (n=9), a group receiving 300 mg/kg ARQ 197 (n=9), were administered orally by gavages for 5 days/week and for 4 weeks from day 4th after *i.v.* injection mice were sacrificed on day 32. The numbers of colonies lung surface metastases tumors were counted. In the vehicle group, massive pulmonary metastasis were formed in all of mice. Treatment with ARQ 197 significantly reduced or prevented lung metastasis, as measured by number of lung surface nodules. These data are illustrated in Fig. A, B.

A

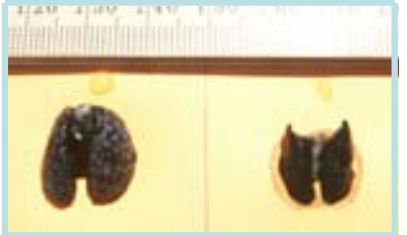

Vehicle 10ml/kg, po. qd | **ARQ 197 300mg/kg, po. qd**

B

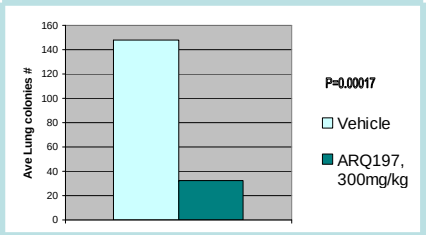

Ave Lung colonies #
160
140
120
100
80
60
40
20
0
P=0.00017
Vehicle
ARQ197, 300mg/kg

Effect of ARQ 197 on experimental metastases following intravenous injection . SCID mice bolus *i.v.* HT29 cells

(9 mice /group) received vehicle, ARQ 197 300mg/kg, po.

A. Representative in situ photographs of lungs from vehicle and ARQ 197 treated mice.

B. The statistical data of average number of lung surface tumor colonies.

Summary

- **These studies demonstrate that the two model system are excellent systems for studying the metastatic behavior of human tumors and for studying the antimetastatic activity of experimental drugs.**
- **ARQ 197 displayed potent activity against cancer metastasis and invasiveness in experimental metastatic models of colon cancer.**
- **The doses and dosing regimens of ARQ 197 used in these murine models of colorectal cancer were well tolerated.**
- **These results support the therapeutic potential of targeting c-Met to prevent cancer metastasis.**
- **These findings support the advance of ARQ 197 in phase I clinical evaluation for the treatment of human malignancies.**

Presented at:

- **2007 AACR Annual Meeting**
 April 14-18, 2007
 Los Angeles, CA