Abstract #2216

Broad Spectrum anti-cancer activity of ARQ 197, a highly selective oral c-Met Inhibitor, in multiple xenograft models

Youzhi Li[2], Dongshu Chen[2], Wei Zhou[2], Wei Li[2], Jieti Sun[2], Ella Hyatt[2], Zhiwei Jiang[2], Sapna Makhija[1], David S. Leggett[2], Chiang J. Li[1,2]

[1]ArQule, Inc., 19 Presidential Way, Woburn, MA 01801, [2]Boston Biomedical, Inc., 333 Providence Highway, Norwood, MA 02062

ABSTRACT

The inappropriate expression of c-Met, a receptor tyrosine kinase, in most cancers and its role in controlling multiple signal transduction pathways involved in tumor growth and metastasis render it a compelling therapeutic target for cancer. ArQule has discovered ARQ 197, a small molecule drug that potently and selectively inhibits c-Met. In this study, we have assessed the spectrum of anti-cancer activity *in vivo* by inhibiting c-Met with ARQ 197. Multiple xenograft models including breast cancer, prostate cancer, colon cancer and pancreatic cancer were used in our studies. ARQ 197 was administered orally by gavage for 5 days/week and for 4 weeks. Our results showed that treatment with 200 mg/kg of ARQ 197 significantly blocked the growth of MDA-MB-231 xenografted tumor with a tumor growth inhibition (TGI) of 79% (p=0.009). Similarly, we observed a TGI of 71.5% (p=0.014) at a dose of 200 mg/kg and a TGI of 76.5% (p=0.01) at a dose of 300 mg/kg in a PC3 model. In a HT29 model, treatment with 300 mg/kg of ARQ 197 (TGI was 55%, p=0.0000128) was more effective than with 200 mg/kg (TGI was 39%, p=0.006). In a Paca2 model, treatments at either 300 mg/kg or 200 mg/kg resulted in a TGI of about 60% (p values were 0.018 or 0.036, respectively). No adverse effects were observed based on general appearance, weight loss or reduction in weight gain. In summary, ARQ 197, a highly selective oral c-Met inhibitor, exhibits broad spectrum anti-tumor activity *in vivo*, supporting c-Met as a highly promising therapeutic target against cancer.

Introduction

c-Met is a transmembrane tyrosine kinase receptor for Hepatocyte growth factor/scatter factor (HGF/SF) encoded by the *c-met* proto-oncogene. Its role in cancer development is definitively established as it is inappropriately expressed in many types of human cancer. It plays key roles in controlling multiple signal transduction pathways involved in cell growth, cell survival, angiogenesis, invasion and metastasis. Therefore, c-Met is a highly compelling therapeutic target for human cancer.

ARQ 197 has shown a broad-spectrum activity against cancer cells and selectively and potently inhibits c-Met. In this report, we describe the preclinical pharmacological profile of ARQ 197 including the anti-tumor effects and safety of ARQ 197 with multiple xenograft models in mice. ARQ 197 demonstrated broad acting preclinical antitumor efficacy and a tolerability profile amenable to chronic p.o. administration. Based on these findings, ARQ 197 has been advanced to clinical trials.

Materials & Methods

Animals.

All experiments were carried out using female or male 6-8 week old athymic nude mice (Charles River Laboratory, Wilmington, MA). Animals were maintained in microisolation cages and maintained in accordance with the guiding principles of ArQule's Institutional Animal Care and Use Committee throughout the studies.

Injection of Cell Lines.

The MDA-MB-231, PC3, HT29 and Mia-Paca-2 cell lines were obtained directly from the ATCC catalogue. Approximately 5-8x 10^6 cells were injected subcutaneously into the right flank of each animal. When tumors reached a volume of ~100 mm^3, animals were randomized into treatment groups (n=8-10).

Drug Administration.

The prepared test and control substances were administered via oral administration with either vehicle control or ARQ 197 formulated in PEG400:20% Vitamin E TPGS (60:40) at 30 mg/ml. treated daily (5 consecutive days, followed by a 2-day dosing holiday) for four week period.

Tumor Measurements.

Tumors were measured two times per week during the treatment and observation periods. Calipers were used to measure each tumor at its longest axis and its shortest dimension. Tumor volume was calculated using the following equation: (length x (width)2)/2 and expressed in mm3.

Tumor growth inhibition.

Mean tumor growth inhibition of each treated group was compared with vehicle control and a TGI value calculated using the formula:

[1-(T/C) x100%].

Statistical analyses.

Statistical analyses in xenograft studies were performed comparing test agent to control groups using a two-tailed Student t-test with P<0.05 deemed significant.

RESULTS

The antitumor efficacy and tolerability of chronic p.o. ARQ 197 administration were investigated in a therapeutic context against a series of established human tumor xenografts implanted s.c. in athymic nude mice. These data are illustrated in Figs.1-4. ARQ 197 was administered orally by gavages for 5 days/week and for 4 weeks. This regimen resulted in dose-related growth inhibition of multiple human s.c. tumor xenografts ranging from 50-80% including breast cancer, prostate cancer, colon cancer and pancreatic cancer.

1. Breast Cancer Xenograft MDA-MB-231 Model

A B

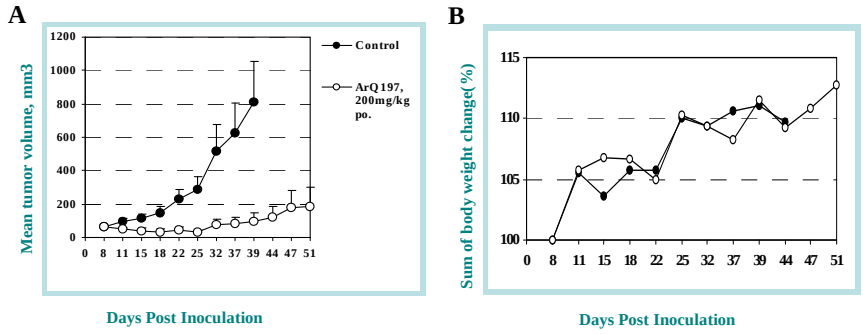

Mean tumor volume, mm3
Control
ArQ197, 200mg/kg po.
Days Post Inoculation
Sum of body weight change(%)
Days Post Inoculation

Figure 1.

A:Antitumor activity in a human breast cancer xenograft MDA-MB-231 model.
Immunosuppressed mice with established subcutaneous MDA-MB-231 human breast cancer were given ARQ 197 (200 mg/kg), or vehicle control orally. All regimens were administered daily. Tumor size was evaluated periodically during treatment. Each point represents the arithmetic mean ± SE of ten tumors.

B: The whole body weight was measured periodically during treatment.

2. Prostate Cancer Xenograft PC3 Model

A B

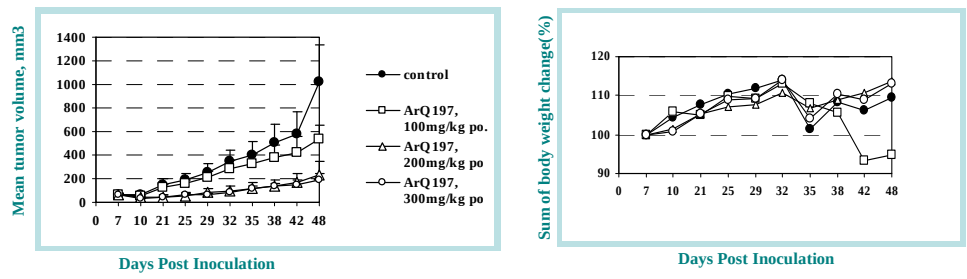

Mean tumor volume, mm3
control
ArQ197, 100mg/kg po.
ArQ197, 200mg/kg po
ArQ197, 300mg/kg po
Days Post Inoculation
Sum of body weight change(%)
Days Post Inoculation

Figure 2.

A:Antitumor activity in a human prostate cancer xenograft PC3 model.
Immunosuppressed mice with established subcutaneous PC3 human prostate cancer were given ARQ 197 (100, 200 and 300mg/kg), or vehicle control orally. All regimens were administered daily. Tumor size was evaluated periodically during treatment. Each point represents the arithmetic mean ± SE of ten tumors.

B: The whole body weight was measured periodically during treatment.

3. Colon Cancer Xenograft HT29 Model

A B

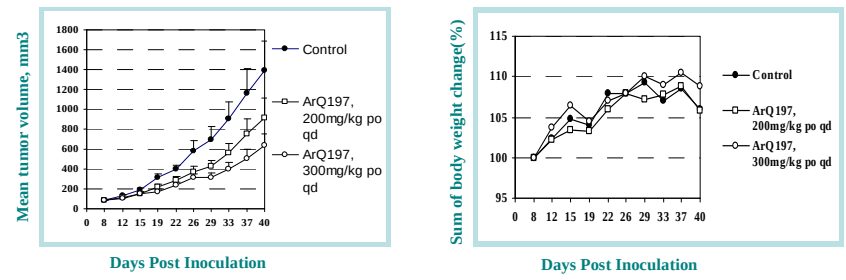

Mean tumor volume, mm3
Control
ArQ197, 200mg/kg po qd
ArQ197, 300mg/kg po qd
Days Post Inoculation
Sum of body weight change(%)
Control
ArQ197, 200mg/kg po qd
ArQ197, 300mg/kg po qd
Days Post Inoculation

Figure 3.

A: Antitumor activity in a human colon cancer xenograft HT29 model.
Immunosuppressed mice with established subcutaneous HT29 human colon cancer were given ARQ 197 (200 mg/kg), ARQ 197 (300 mg/kg) or vehicle control orally. All regimens were administered daily. Tumor size was evaluated periodically during treatment. Each point represents the arithmetic mean ± SE of ten tumors.

B: The whole body weight was measured periodically during treatment.

4. Pancreatic Cancer Xenograft Paca-2 Model

A B

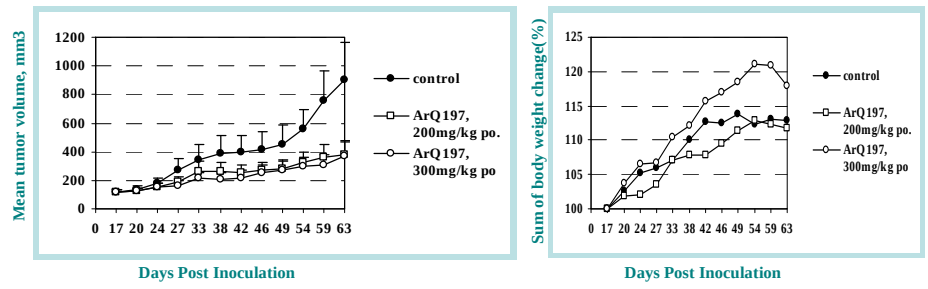

Mean tumor volume, mm3
control
ArQ197, 200mg/kg po.
ArQ197, 300mg/kg po
Days Post Inoculation
Sum of body weight change(%)
control
ArQ197, 200mg/kg po.
ArQ197, 300mg/kg po
Days Post Inoculation

Figure 4.

A: **Antitumor activity in a human pancreatic cancer xenograft Paca-2 model**.
Immunosuppressed mice with established subcutaneous Paca-2 human pancreatic cancer were given ARQ 197 (200 mg/kg), ARQ 197 (300 mg/kg) or vehicle control orally. All regimens were administered daily. Tumor size was evaluated periodically during treatment. Each point represents the arithmetic mean ± SE of ten tumors.

B: **The whole body weight was measured periodically during treatment**.

Summary

- **ARQ 197 displayed significant anti-tumor activity in multiple tumor xenograft models.**
- **ARQ 197 is an orally bioavailable, well-tolerated cMet inhibitor**.
- **These results support the therapeutic potential of targeting c-Met in cancers where c-Met plays a role in tumor growth or metastasis**
- **These findings support ARQ 197 as an exciting and promising candidate currently in phase I clinical evaluation for the treatment of human malignancies.**

Presented at:

- **2007 AACR Annual Meeting**
 April 14-18, 2007
 Los Angeles, CA